The Immunoglobulin Diagnosis, Evaluation, and key Learnings (IDEaL) Patient Registry: Baseline Clinical Assessment and Outcomes Results from Our Primary Immune Deficiency Population

Sean Kearns, PhD,[1] Loretta Kristofek, RN, BSN,[1] Bill Bolgar, PharmD,[1] and Luqman Seidu, MD[2]

[1]Coram Clinical Trials, Denver, CO | [2]Omni Allergy, Immunology and Asthma, Atlanta, GA

Introduction

Long-term outcomes and patient-reported quality-of-life assessments are not widely available for patients on immunoglobulin (Ig) therapy for different disease types. The IDEaL Patient Registry collects longitudinal information on subjects receiving Ig replacement therapy from Coram CVS/specialty infusion services in an alternate care setting, primarily in the home. Patients from our 140 investigators are eligible to enroll in the Registry. For this study, we examined the IDEaL primary immune deficiency (PID) patient population, reviewing data for treated patients including baseline lab values as well as infection rates and quality-of-life assessments over time.

Methods

IRB approval was acquired, and informed consent was obtained from all subjects. Information collected by Coram nurses and pharmacists was entered into the IDEaL database. Additionally, subjects were asked to complete an SF-36 questionnaire and a Life Quality Index Questionnaire (LQIQ) every six months.

Results

Currently 353 subjects are enrolled in the Registry, and over 85% of these subjects (n=301) are being treated with IgG for PID. The most common diagnosis among these patients is common variable immunodeficiency (CVID); approximately 78% of our PID patients have this diagnosis.

Patient Demographics

Figure 1 shows the breakdown of gender and age for PID patients in the Registry. In the pediatric population, there were slightly more males enrolled, as compared to the "18 to 64 years" and "65 years and over" age brackets, in which enrolled females significantly outnumbered males. The average ages for each category are listed, with the overall pediatric age at time of admission being nine years of age, and the average age of adults at time of admission being 59 years of age.

Figure 1. Gender and Average Age Breakdown of PID Patients

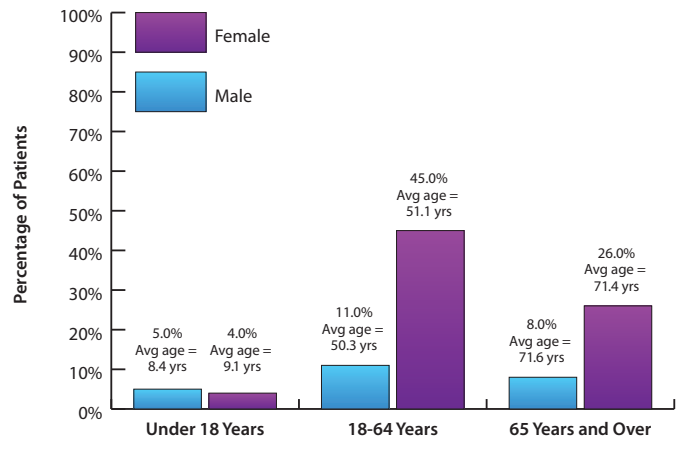
Female
Male
Percentage of Patients
5.0% Avg age = 8.4 yrs
4.0% Avg age = 9.1 yrs
11.0% Avg age = 50.3 yrs
45.0% Avg age = 51.1 yrs
8.0% Avg age = 71.6 yrs
26.0% Avg age = 71.4 yrs
Under 18 Years
18-64 Years
65 Years and Over

Figure 2a. IgG Serum Level Distribution

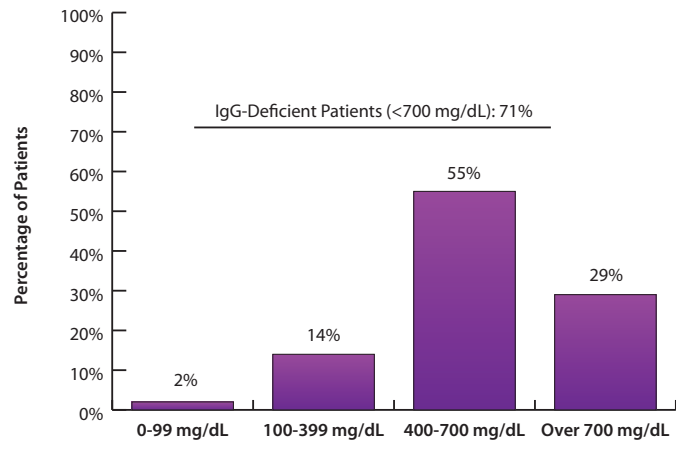
Percentage of Patients
IgG-Deficient Patients (<700 mg/dL): 71%
2%
14%
55%
29%
0-99 mg/dL
100-399 mg/dL
400-700 mg/dL
Over 700 mg/dL

IgG Serum and Subclass Deficiencies

Figure 2A shows the distribution of subject baseline serum IgG levels. Out of 128 patients, 71% (n=91) showed a deficiency in serum IgG level, defined as <700 mg/dL. The majority of these patients had levels between 400–700 mg/dL (55% of all patients), a range considered to represent a mild or moderate decline in IgG. The overall average IgG level for patient values flagged as being below the reference range minimum was 509 mg/dL.

IgG subclass deficiencies can contribute to declines in immune function, even in patients with a normal total serum IgG level. Figure 2b shows the breakdown of serum and subclass deficiencies in the PID patient population within the Registry. Out of 161 patients, 47 patients had only serum IgG levels analyzed; no subclass levels were analyzed. In patients with low serum IgG levels whose subclasses were tested (n=76), the majority showed declines in two or more subclasses (mainly IgG1 and IgG2 [25%]) and in three or more subclasses (IgG1+2+4; 17%). In patients with serum IgG levels in the normal range and with specific subclass deficiency (n=38), the majority showed declines in IgG1 (18%) and IgG2 (42%), with no patient having more than two subclass deficiencies.

Figure 2b. IgG Serum and Sublcass Distribution

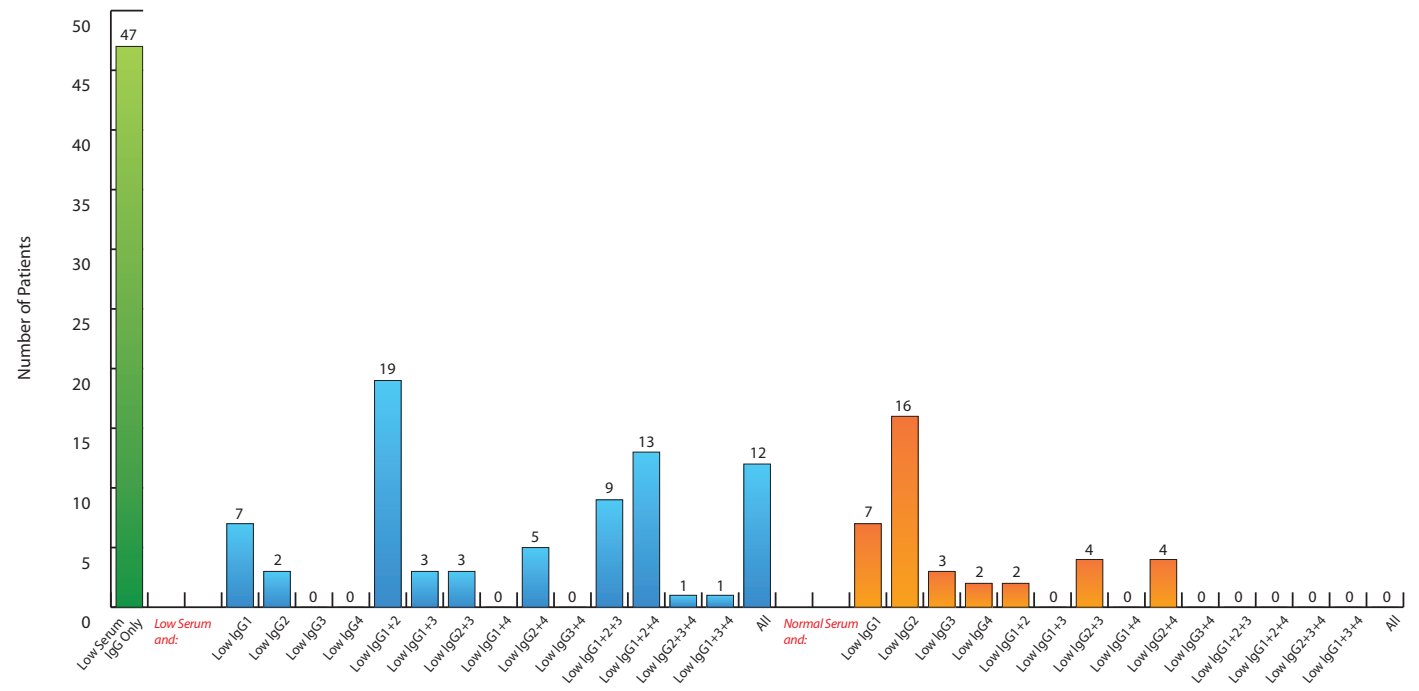
Number of Patients
Low Serum IgG Only
Low Serum and:
Normal Serum and:

Pneumococcal Vaccine Response

Pneumococcal vaccine challenge is a diagnostic test commonly used to assess immune system function. In our PID patient population, we analyzed the percentage of serotypes to which subjects responded (Figure 3). Of all subjects, 49% responded to 25% or less of the tested serotypes, and a clear majority (75%) responded to half or less of the tested serotypes.

Figure 3. Pneumococcal Vaccine: Percentage of Serotypes with a Positive Response (>1.3 ug/mL)

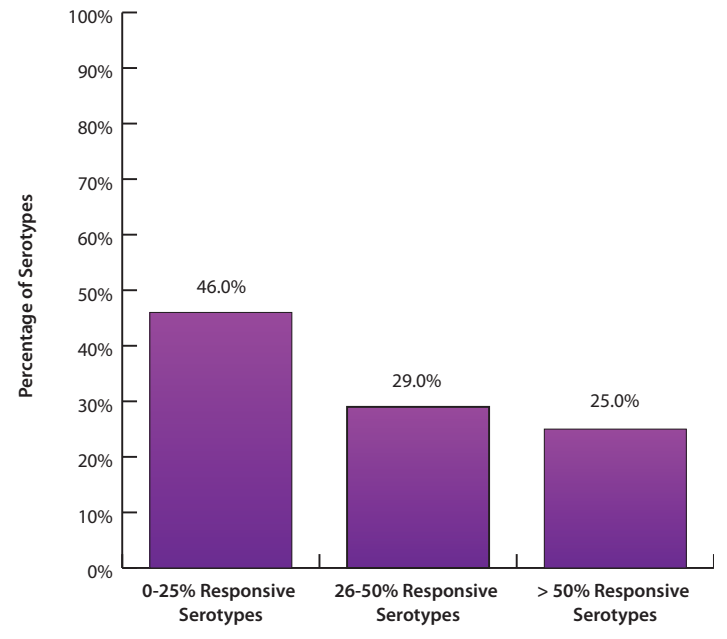
Percentage of Serotypes
46.0%
29.0%
25.0%
0-25% Responsive Serotypes
26-50% Responsive Serotypes
> 50% Responsive Serotypes

We also examined the correlation between IgG serum levels and pneumococcal vaccine response (Figure 4). We saw a weak correlation between serum IgG levels and pneumococcal response, but did note that in those subjects who had normal serum IgG levels (>700 ug/dL) and showed a response to half or less of the pneumococcal serotypes, 67% of them had a low IgG2 level. This is suggestive that IgG2 may play a larger role in pneumococcal response than the other subclasses, and subjects with low IgG2 (even with normal serum levels) may be more susceptible to sinus and upper respiratory infections.

Ig Administration, Dosing, and Infection Response

For PID subjects, both intravenous and subcutaneous routes of Ig administration are available. The average dose for subcutaneous Ig (SCIg) was 536 mg/kg/month (equal to 134 mg/kg/week); for intravenous Ig (IVIg), it was 465 mg/kg/month (Figure 5).

Baseline and Trough Serum IgG Level

Patients were split according to the route of their Ig administration, and baseline and trough IgG levels were compared. At baseline, patients getting IVIg averaged a serum IgG level of 582 mg/dL, and patients on SCIg averaged 609 mg/dL. IVIg trough levels averaged 1,018 mg/dL and SCIg troughs averaged 1,120 mg/dL (Figure 5a). These values suggest that regardless of route, patients are being held at IgG levels that fall right in the middle of the normal reference ranges.

The average annual number of infections was three, with no significant difference between administration routes. Figure 6A shows the dose response plot for SCIg infusions and average annual rates of breakthrough infections; Figure 6B shows the plot for IVIg.

Figure 4. Correlation Between Serum IgG Levels and Pneumococcal Vaccine Response

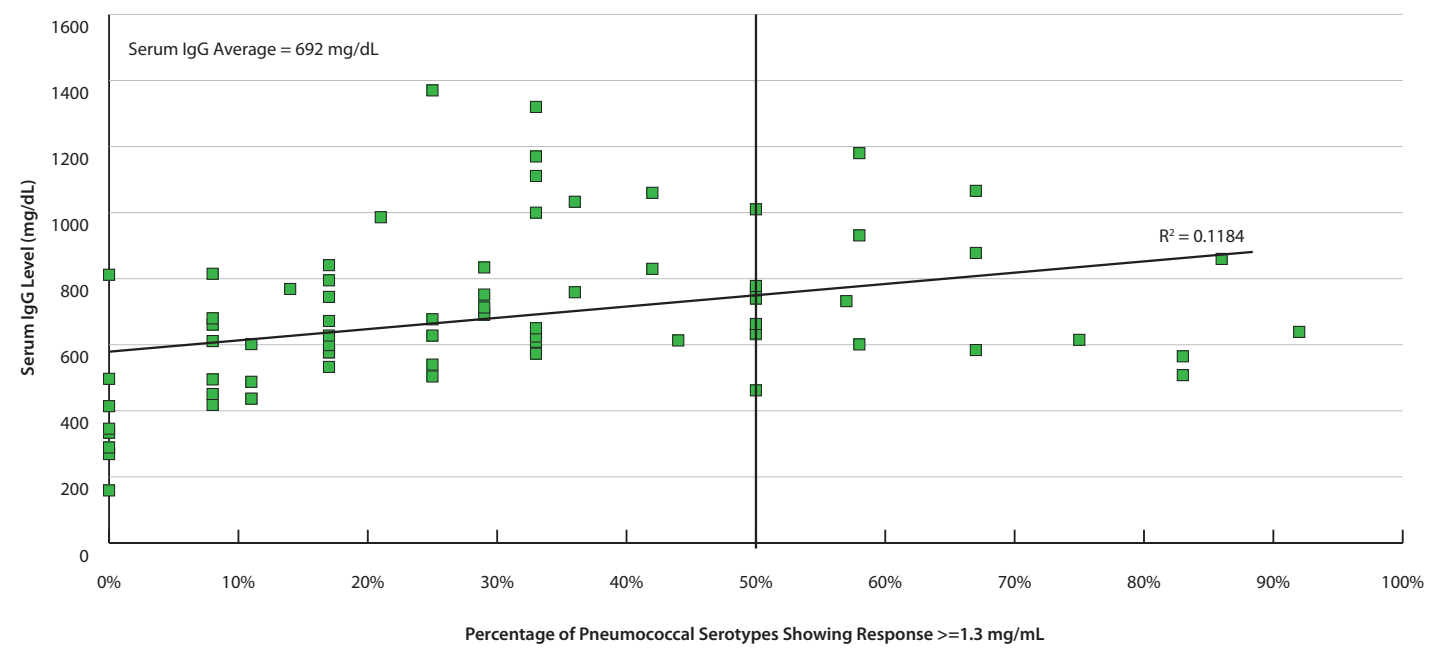
Serum IgG Average = 692 mg/dL
R^2 = 0.1184
Serum IgG Level (mg/dL)
Percentage of Pneumococcal Serotypes Showing Response >=1.3 mg/mL

Figure 5. Monthly IgG Dosing by Route

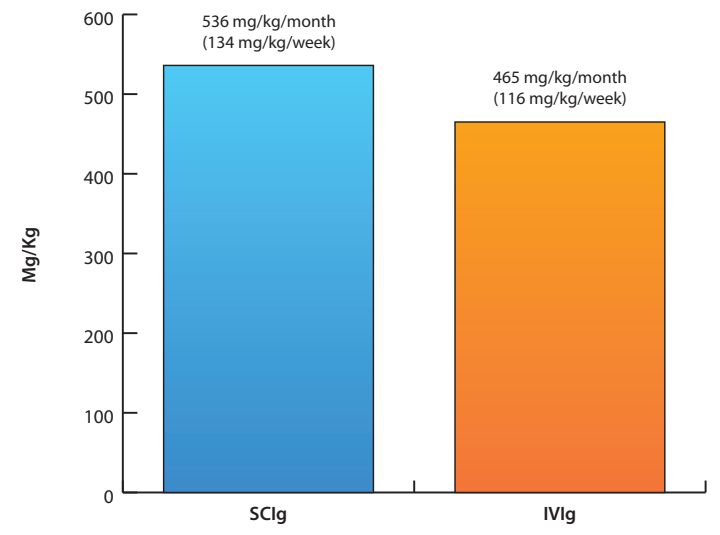
Mg/Kg
536 mg/kg/month (134 mg/kg/week)
465 mg/kg/month (116 mg/kg/week)
SCIg
IVIg

Figure 5a. Serum IgG Levels: Baseline and Trough Values

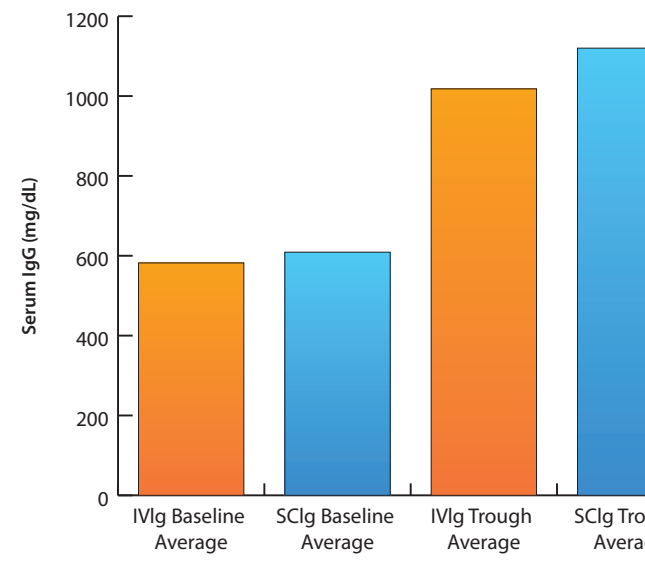
Serum IgG (mg/dL)
IVIg Baseline Average
SCIg Baseline Average
IVIg Trough Average
SCIg Trough Average

Figure 6a. SCIg Dose and Infection Response

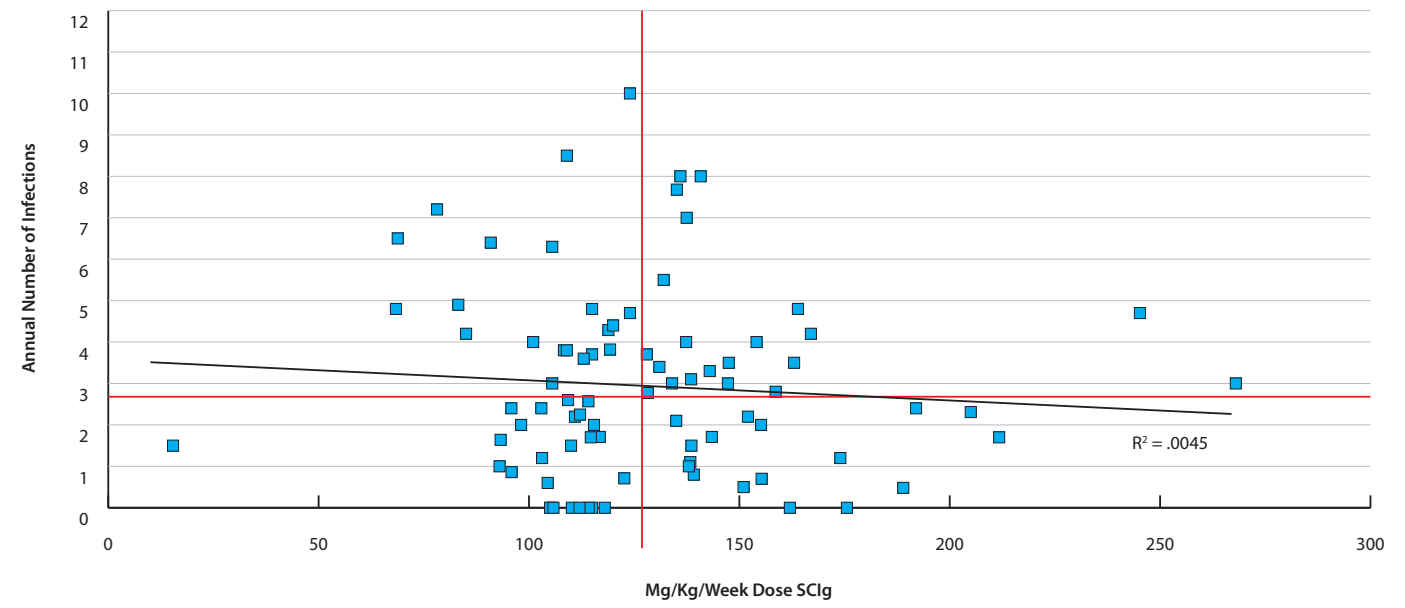
Annual Number of Infections
R^2 = .0045
Mg/Kg/Week Dose SCIg

Figure 6b. IVIg Dose and Infection Response

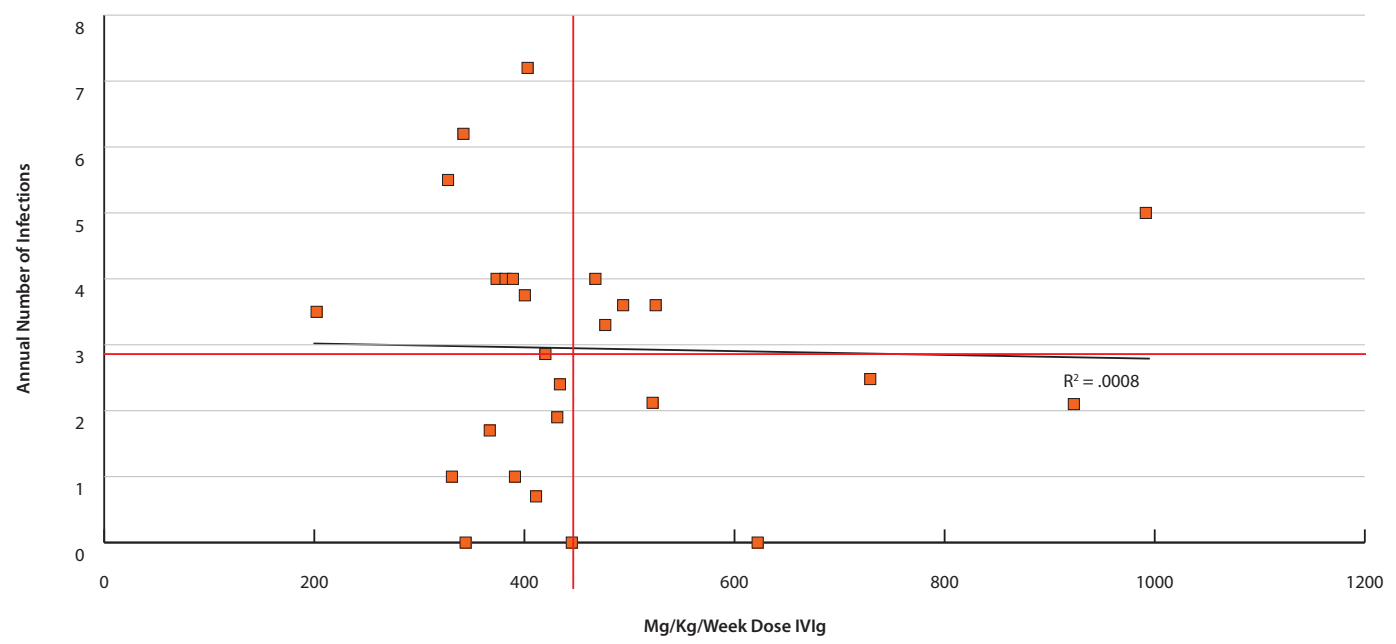
Annual Number of Infections
R^2 = .0008
Mg/Kg/Week Dose IVIg

Health Transition Index — Symptom Change Over Time

The SF-36 Health Questionnaire allows for longitudinal tracking of patients' health over the course of their treatment. Patients are asked to rate their overall health, as compared to one year prior. We examined their responses at the 6-, 12-, and 24-month time points to gauge how their treatments had impacted their health as compared to their responses before treatment, at the start of treatment, and after a while on treatment. At all time points and for both routes of administration, we noted that a majority of patients stated that their health had improved or stayed the same (Figure 7).

Figure 7. SF-36 Health Transition

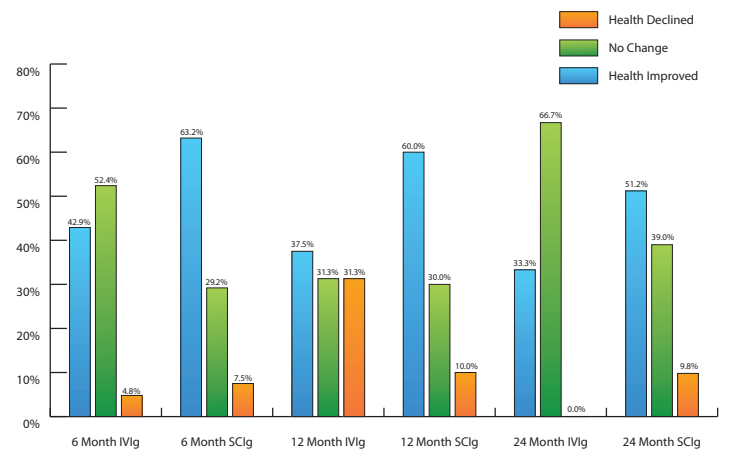
Health Declined
No Change
Health Improved
6 Month IVIg
6 Month SCIg
12 Month IVIg
12 Month SCIg
24 Month IVIg
24 Month SCIg

Conclusions

These results from the IDEaL Registry suggest an overall profile of PID patients receiving Ig. Key findings include:

- Adult subjects were diagnosed later in life, with an average age at diagnosis of 59 years, while pediatric subjects were diagnosed on average around age 9 years.
- The majority of our subject population had baseline IgG levels between 400–700 ug/dL, and a sizeable minority had normal serum IgG levels but was deficient in one or more subclasses.
- There is a weak correlation between serum IgG levels and pneumococcal vaccine response, but in subjects with normal serum IgG and responsiveness to half or less of the pneumococcal serotypes, a significant majority was deficient in IgG2.
- Our average SCIg dose was toward the low end of the recommended dose range, but both IVIg and SCIg patients showed trough levels in the middle of the normal reference range and greater than 1,000 ug/mL.
- Subjects appeared to have good infection control; however, with a limited dose spread, we could not demonstrate a strong dose response curve.
- Subjects generally had a good perception of their treatment and its impact on their health and quality of life. When asked to compare their health now to a time immediately before or at the start of their treatment, the majority of patients noted either no decline or an improvement in their overall health.

Continued data collection will allow for further long-term data analysis on outcomes of patients on Ig treatment, and further contributions to best practices surrounding IgG administration for primary immune deficiencies.